First-in-human study of acoustic cluster therapy (ACT) consisting of PS101 with chemotherapy and insonation in patients with liver metastases of colorectal cancer origin (NCT04021277)

Abstract 6221

Udai Banerji [1,2], Wing Yau [2], Mark O'Leary [2,1], Nigel Bush [1] Sumita Gurung [2], Amir Snapir [3], Melina Mühlenpfordt [3], Jeff Bamber [1,2] and Nina Tunariu [1,2]
[1] The Institute of Cancer Research [2] The Royal Marsden Hospital NHS Foundation Trust, London, United Kingdom; [3] Exact Therapeutics AS, Oslo, Norway

Background

PFB μ-bubble PFMCP μ-droplet

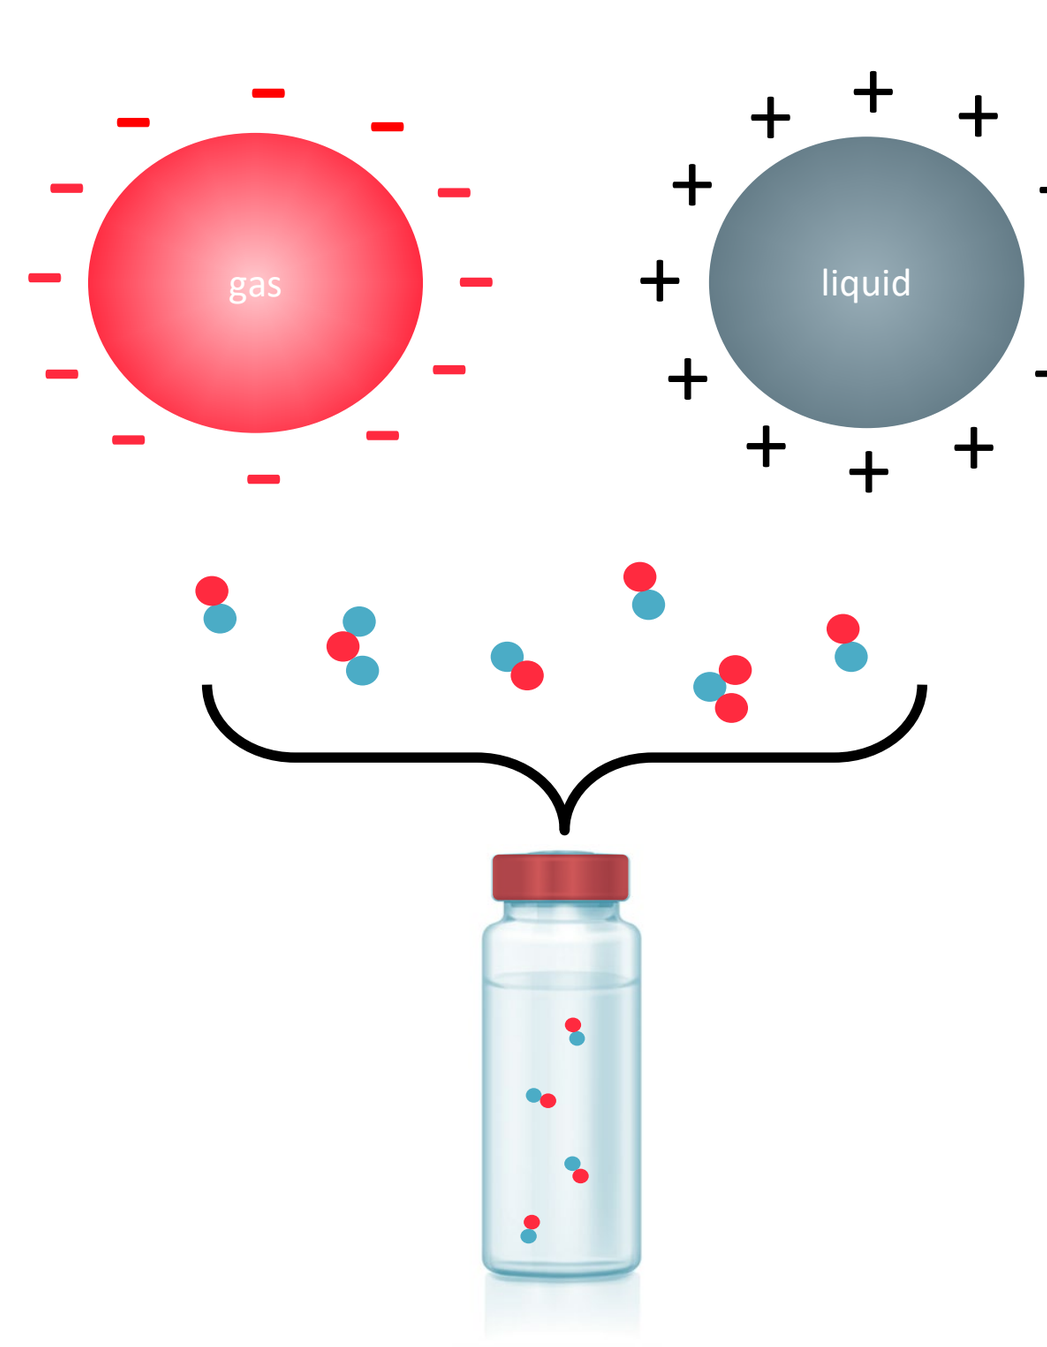
gas
liquid

PS101

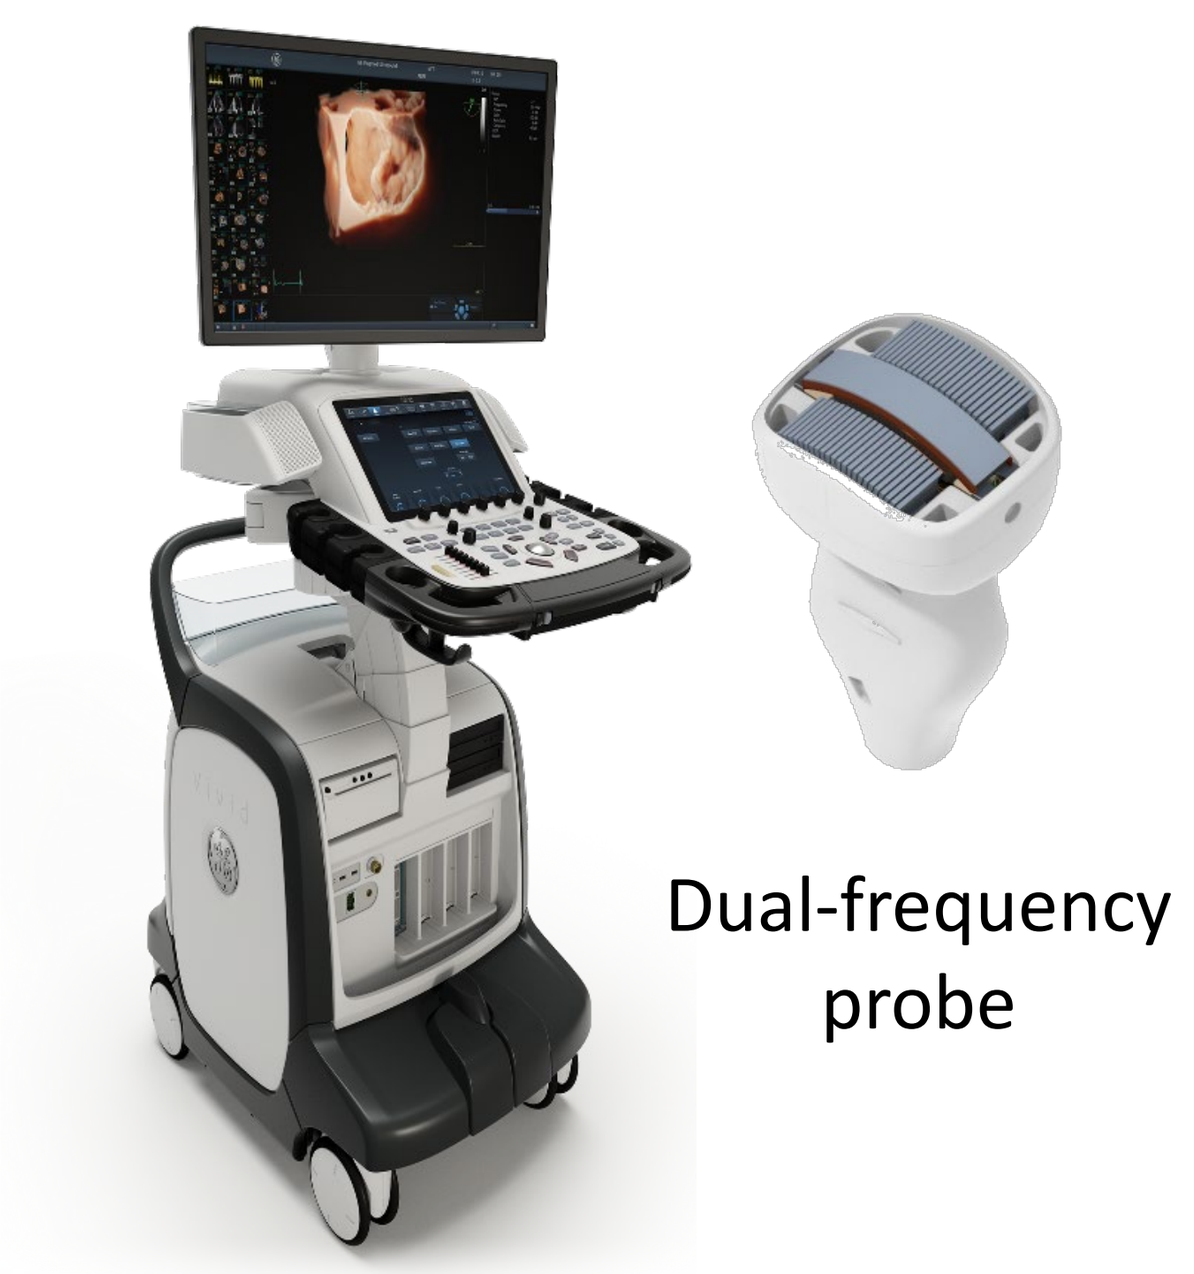

Software modified GE Vivid™ E95

Dual-frequency probe

Step 1: Activation with high frequency (HF) 2-10MHz

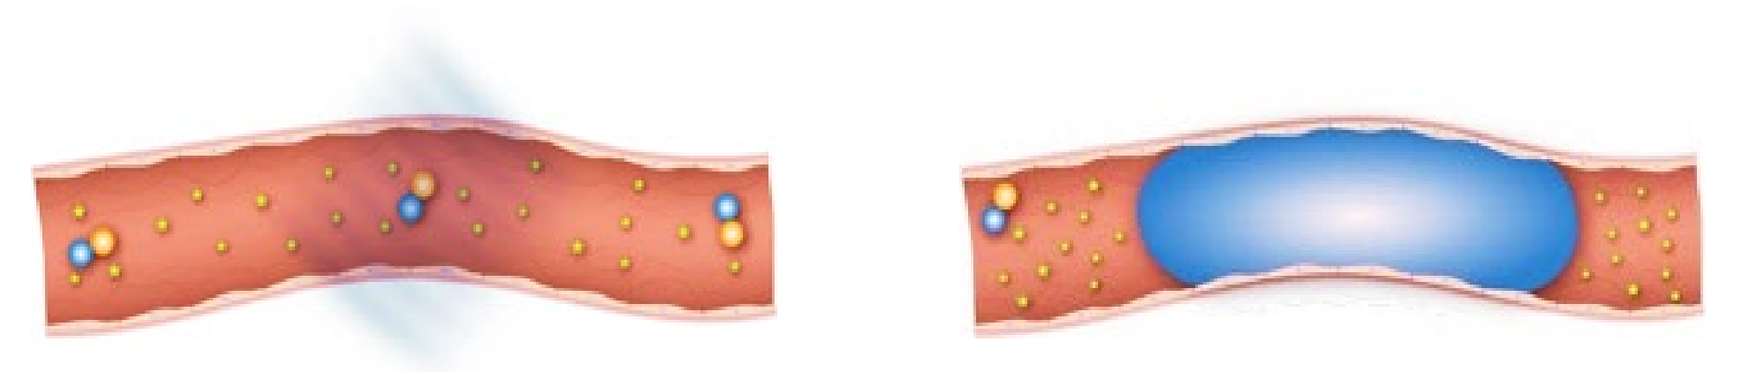

Microclusters (PS101) are free floating in the capillary and HF ultrasound is applied for Activation.

Phase shift and ACT® bubble formation as a result of ultrasound activation.

Step 2: Enhancement with low frequency (LF) 0.5MHz

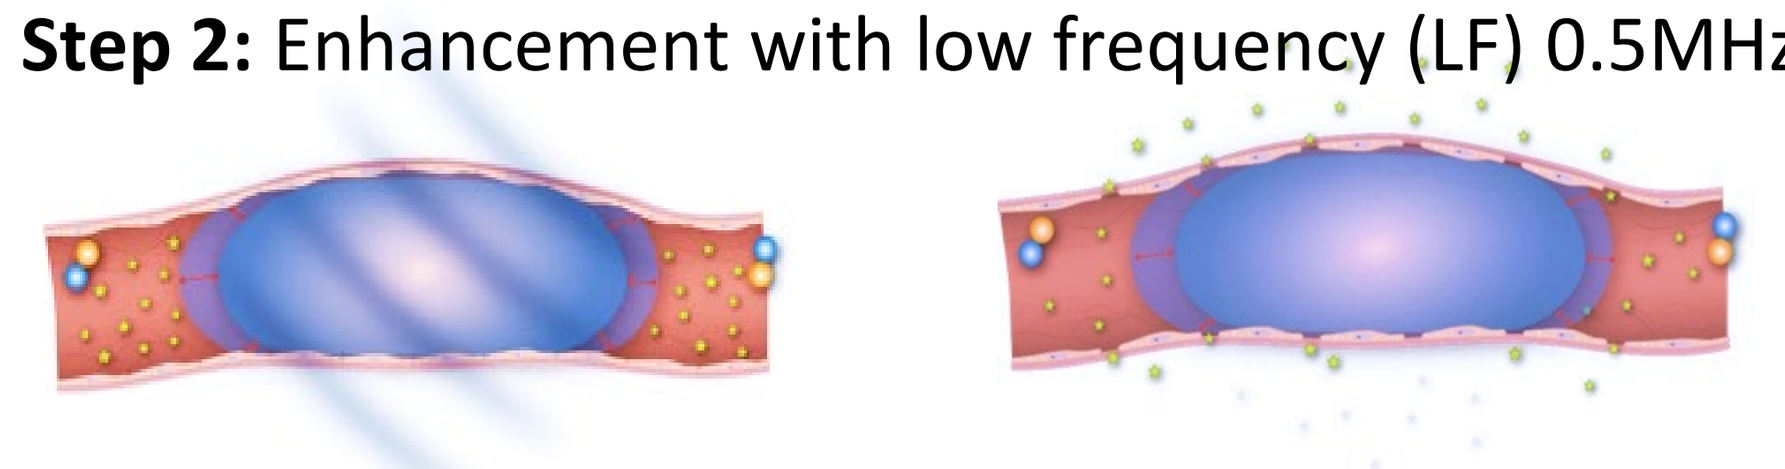

LF ultrasound is applied to the trapped ACT® bubble resulting in controlled oscillation.

Opening of vasculature, extravasation of co-administered drug resulting in Enhanced drug delivery.

Preclinical Experiments

Methods

- Human colorectal adenocarcinoma (SW620) xenografts were implanted subcutaneously into athymic nude mice
- Mice were enrolled into the study when tumour sizes reached ~90 mm^3
- The therapeutic efficacy of ACT with irinotecan (60 mg/kg i.p.) was investigated using three treatment sessions given on day 0, 7, and 14
- ACT treatment was performed with three back-to-back PS101 injections on each treatment day using 45 sec of high frequency ultrasound at 8 MHz at a mechanical index of 0.3 and low frequency exposure at 500 kHz with a mechanical index of 0.2

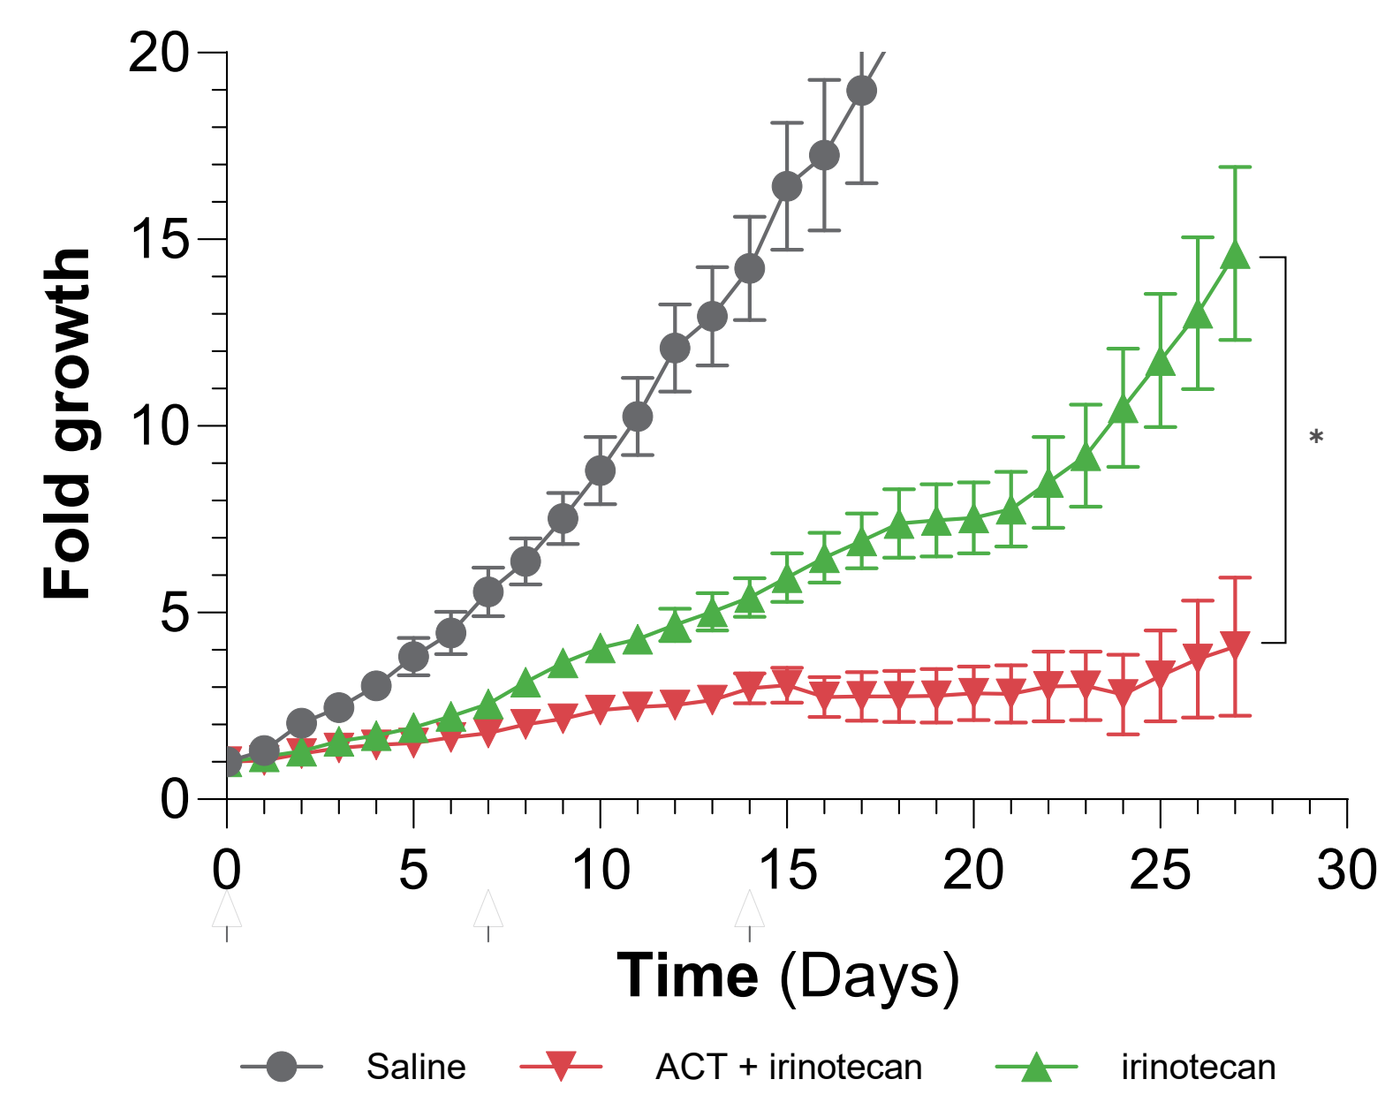
Fold growth
20
15
10
5
0
0
5
10
15
20
25
30
Time (Days)
*
Saline
ACT + irinotecan
irinotecan

Results

- ACT induced a reduction in tumour volume from 14.6 (irinotecan), to 5.4 (ACT with irinotecan, p = 0.002)
- Median survival increased from 34 days (irinotecan) to 54 (ACT with irinotecan, p = 0.002)
- ACT with irinotecan induced an increase in the fraction of complete responders; from 7% to 26%

Conclusion

- ACT with irinotecan for the treatment of colorectal cancer showed a significant improvement in treatment response and survival

Methods

Study treatment:

- up to 4 cycles of ACT treatment (3x IV bolus of PS101 + ultrasound) given concomitantly with FOLFOX (5-fluorouracil [5FU], leucovorin, oxaliplatin) or FOLFIRI (5-FU, leucovorin, irinotecan)
- Radiological evaluation of response based on Investigator and central assessment of the maximum diameter and volume of selected lesions at baseline and at 8 weeks from the start of treatment,
- % change in tumour diameters and volumes are the sum of measurements at 8 weeks minus baseline divided by baseline and multiplied by 100, calculated for each subject and averaged for the dose group.

Results

Population characteristics, treatment and toxicity

	20 ul/kg (n=3)	40 ul/kg (n = 5)	Overall (n = 8)
Age, years, mean (SD)	60 (17)	62 (5)	61 (10)
ECOG – 1, n (%)	3 (100%)	5 (100%)	8 (100%)
Cancer type – colorectal, n (%)	3 (100%)	5 (100%)	8 (100%)
Years since diagnosis of liver metastasis, mean (SD)	5.6 (2.1)	4.4 (3.1)	
Prior lines of chemotherapies, median	6	4	
Combined with FOLFOX/FOLFIRI, n/n	2/1	1/4	3/5
Dose limiting toxicity	0	0	0

Representative scans of patient receiving ACT

Screening Week 8

Insonated lesion

Control lesion

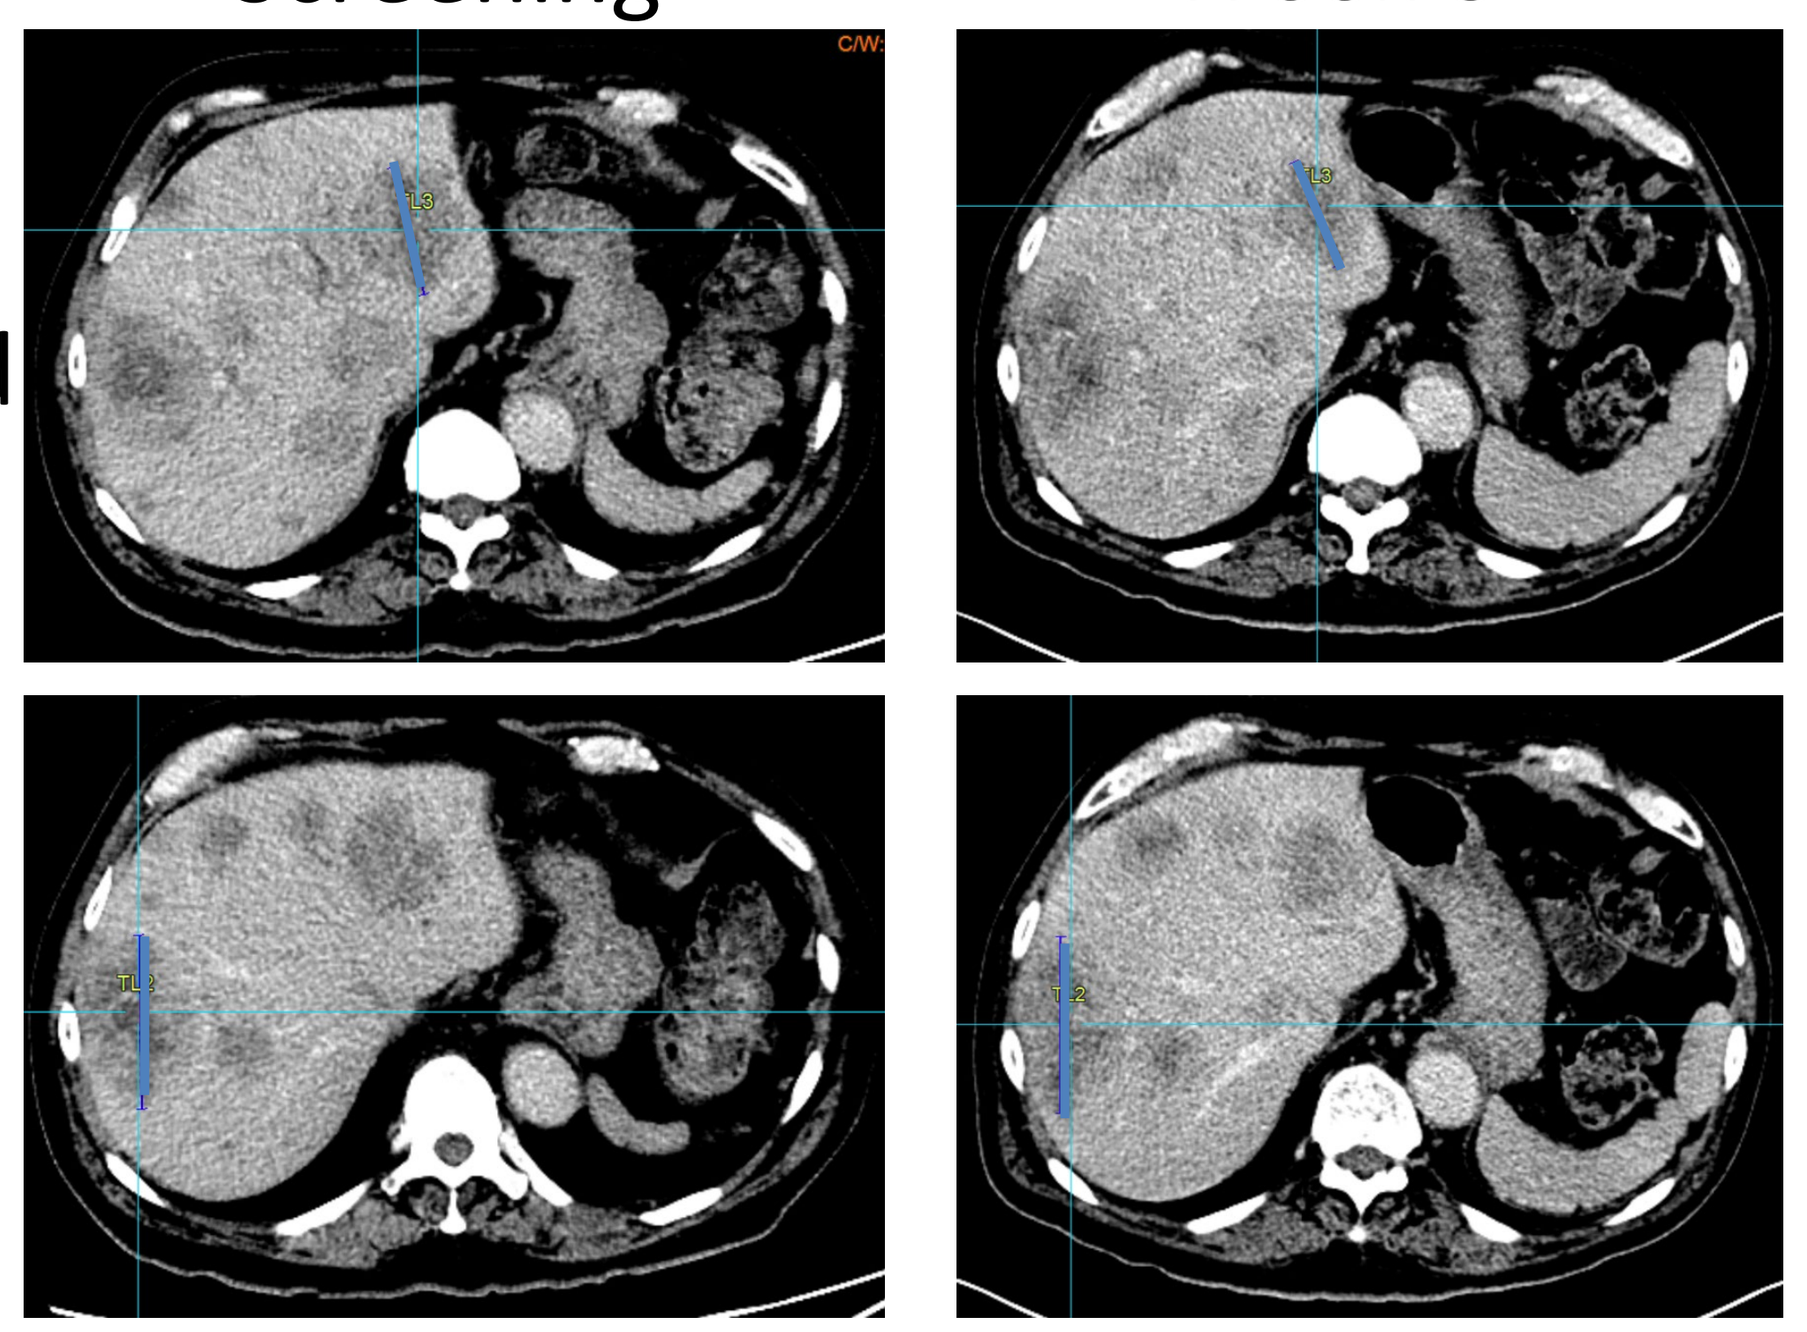

Results

Changes in sum of volumes of control and insonated lesion(s)*

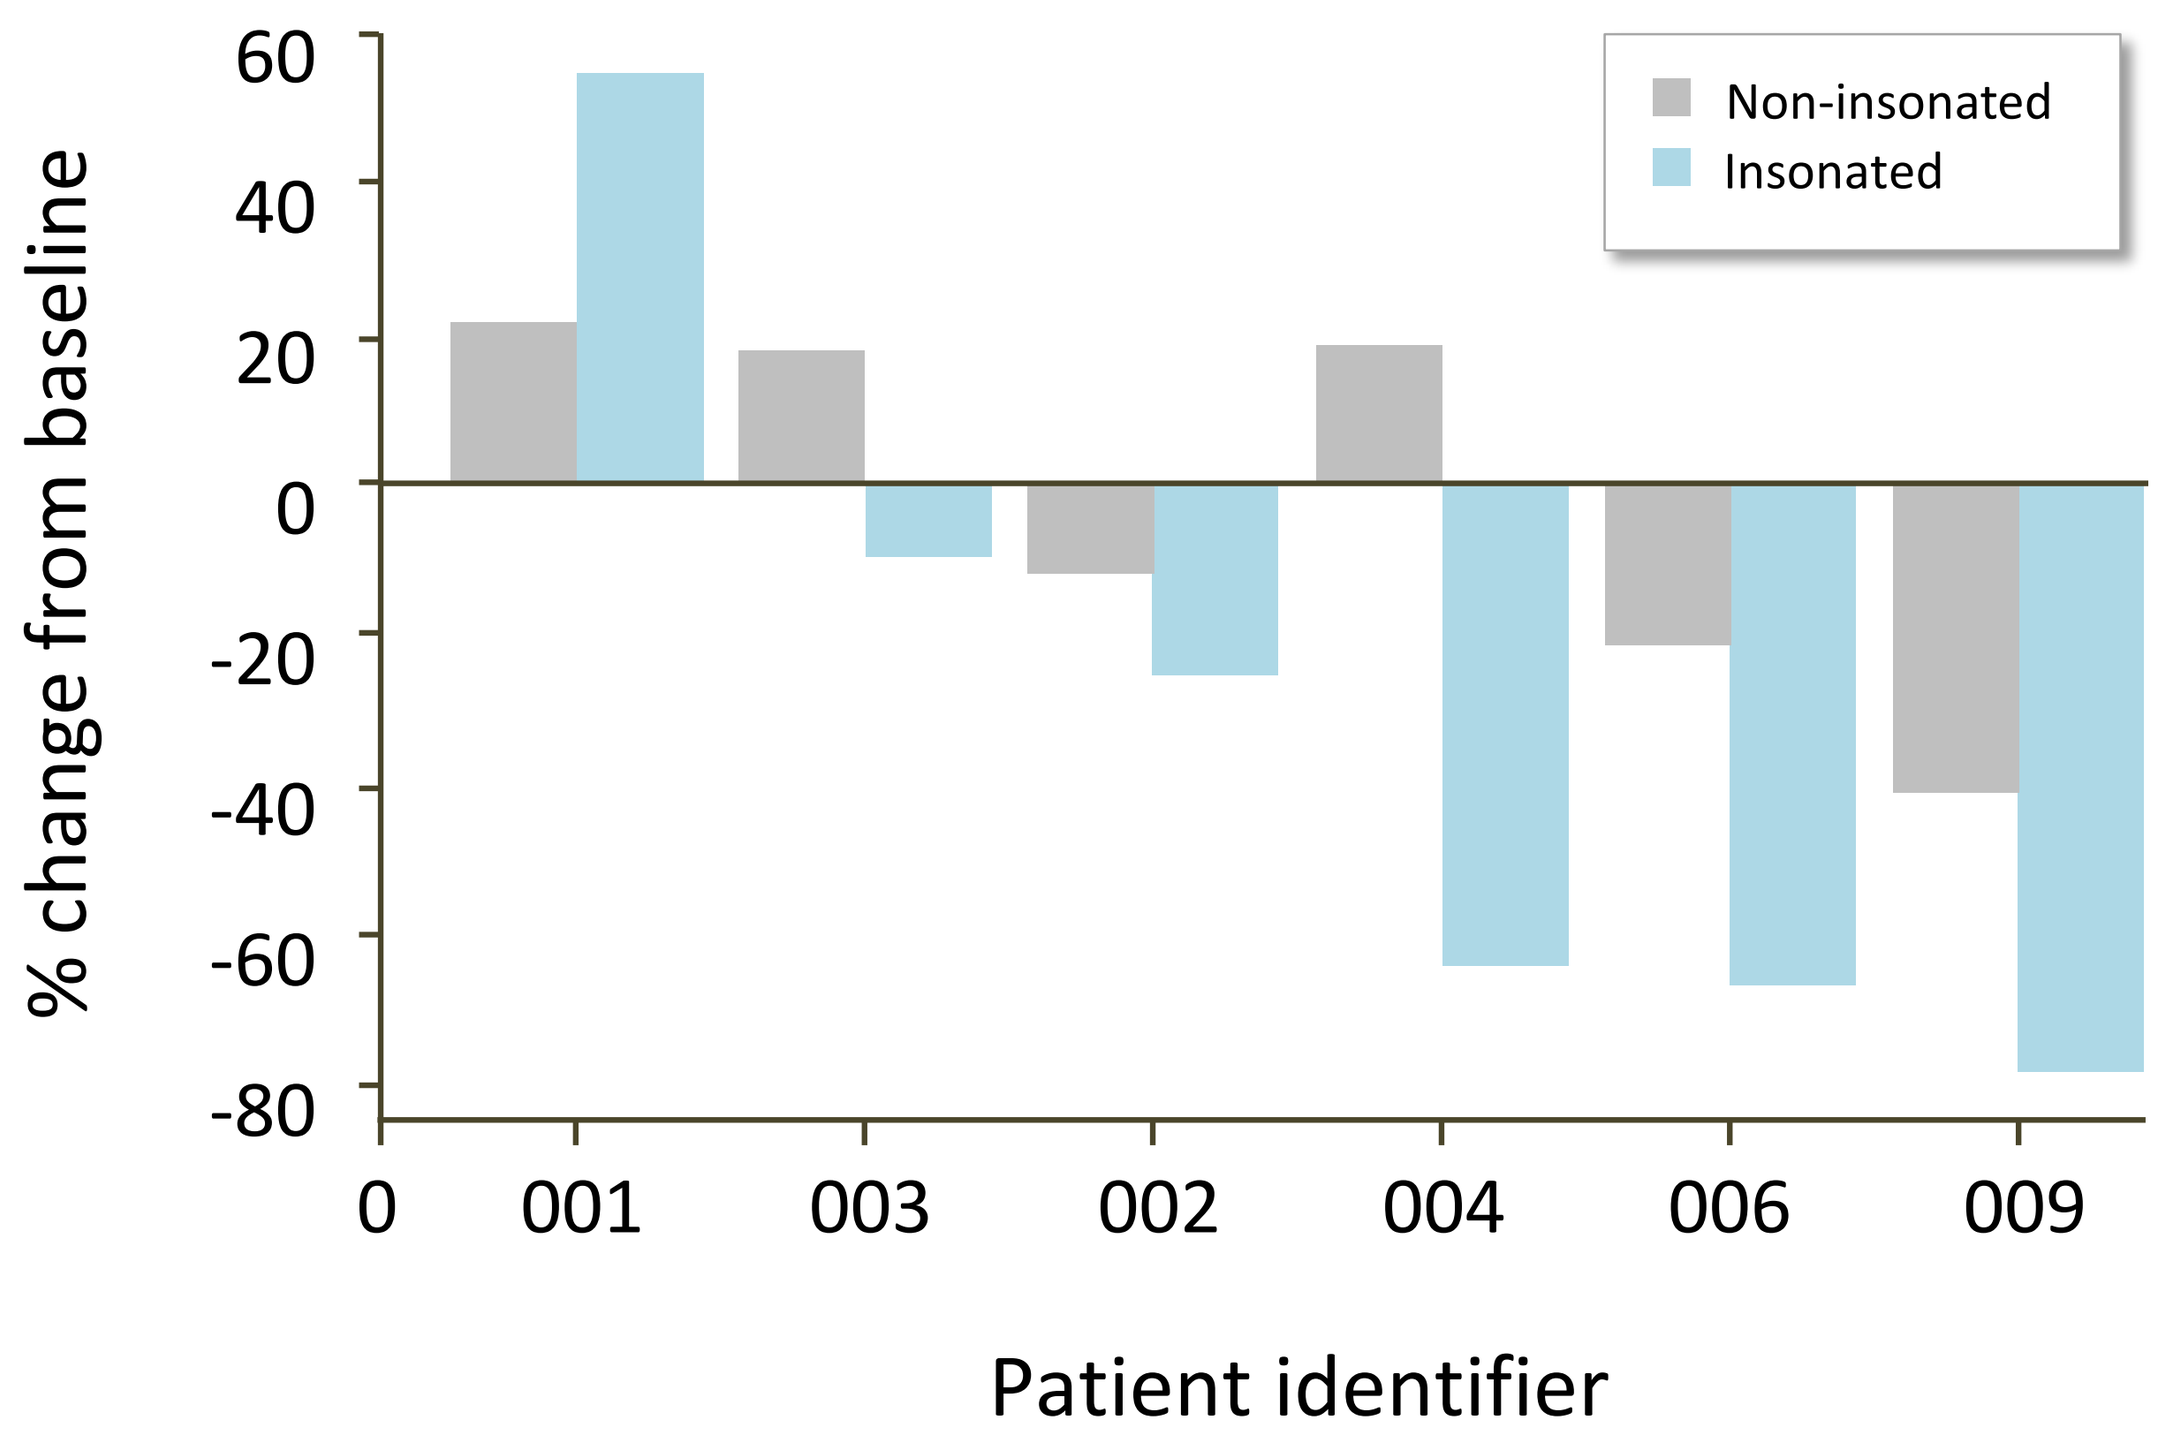
% change from baseline
60
40
20
0
-20
-40
-60
-80
Non-insonated
Insonated
0
001
003
002
004
006
009
Patient identifier

Anti-tumour activity by independent central review
Volume data are average % change from baseline to 8 weeks (SD)

Conclusions

- Data from dose escalation do not show unexpected side effects from PS101 or ultrasound on its own or worsening the effects of chemotherapy
- In the six evaluable patients in the dose escalation cohort, compared to baseline there was a change in volume of -32% (-15% in diameter) in the insonated tumour Vs -3% (-1% in diameter) in the control, non-insonated tumour
- Preliminary data suggests that ACT is deliverable in an out-patient setting and that doses of PS101 are safe and hepatic metastasis can be visualized using microbubbles during chemotherapy
- Data from dose escalation provides proof of concept that insonated lesions show reduction in size of tumour greater than non-insonated lesions. This will be evaluated further in the ongoing expansion part of the ACTIVATE study
- The ACT technology is planned to be evaluated in a Phase 2 study in locally advanced pancreatic cancer - planned to start in 2024

EXACT TX ICR The Institute of Cancer Research The ROYAL MARSDEN NHS Foundation Trust CANCER RESEARCH UK Convergence Science Centre ecmc NIHR Biomedical Research Centre at The Royal Marsden and the ICR

© Poster Template by Genigraphics® 1.800.790.4001 www.genigraphics.com